134P

Intracranial Progression-Free Survival With Ivonescimab Compared to Placebo in the HARMONi-A Trial of Patients With Previously Treated *EGFR* Mutation–Positive Non–Small Cell Lung Cancer

Li Zhang,[1] Wenfeng Fang,[1] Yuanyuan Zhao,[1] Yongzhong Luo,[2] Runxiang Yang,[3] Yan Huang,[1] Zhiyong He,[4] Hui Zhao,[5] Mingjun Li,[6] Kai Li,[7] Qibing Song,[8] Xiaobo Du,[9] Yulan Sun,[10] Wei Li,[11] Fei Xu,[12] Zhiyu Wang,[13] Kunning Yang,[14] Howard (Jack) West,[15] Wenting Li,[16] Michelle Xia[16]

[1]Sun Yat-Sen University Cancer Center, Guangzhou, China; [2]Hunan Center Hospital, Changsha, China; [3]Yunnan Cancer Hospital; The Third Affiliated Hospital of Kunming Medical University; and Yunnan Cancer Center, Kunming, China; [4]Fujian Provincial Tumor Hospital, Fuzhou, China; [5]The Second Hospital of Anhui Medical University, Hefei, China; [6]The first Affiliated Hospital of Zhengzhou University, Zhengzhou, China; [7]Tianjin Medical University Cancer Institute and Hospital, Tianjin, China; [8]Renmin Hospital of Wuhan University, Wuhan, China; [9]Mianyang Central Hospital, Mianyang, China; [10]Shandong Cancer Prevention and Treatment Institute, Jinan, China; [11]The First Affiliated Hospital of Bengbu Medical University, Bengbu, China; [12]First Affiliated Hospital of Nanchang University, Nanchang, China; [13]The Fourth Hospital of Hebei Medical University, Shijiazhuang, China; [14]Weifang Second People's Hospital, Weifang, China; [15]Summit Therapeutics, Menlo Park, CA, USA; [16]Akeso Biopharma Inc., Zhongshan, China

BACKGROUND

- First-line treatment for advanced or metastatic non–small cell lung cancer (NSCLC) with activating epidermal growth factor receptor (*EGFR*) gene variants includes third-generation EGFR tyrosine kinase inhibitors (EGFR TKIs) such as osimertinib[1,2]
- However, NSCLC tumors often develop acquired resistance to EGFR TKIs, which leads to disease progression, leaving patients with limited treatment options[3]
- Ivonescimab is a novel, investigational, bispecific antibody against programmed cell death protein 1 (PD-1) and vascular endothelial growth factor (VEGF); it has exhibited cooperative binding in vitro that could produce complementary and synergistic antitumor effects through the PD-1 and the VEGF pathways[4-6]
- The phase 3 HARMONi-A trial compared the efficacy and safety of ivonescimab plus chemotherapy with chemotherapy in patients with advanced or metastatic *EGFR* mutation–positive NSCLC that progressed after EGFR TKI treatment[7]
 - In the first preplanned interim analysis (March 10, 2023 data cutoff), ivonescimab plus chemotherapy significantly improved progression-free survival (PFS) versus chemotherapy (hazard ratio [HR], 0.46 [95% CI, 0.34-0.62]; *P* < 0.001); median PFS was 7.1 (95% CI, 5.9-8.7) vs 4.8 (95% CI, 4.2-5.6) months[7]
 - The PFS benefit observed with ivonescimab plus chemotherapy compared with chemotherapy was consistent in patients with and without brain metastases at baseline, with HRs of 0.40 (95% CI, 0.22-0.73) and 0.48 (95% CI, 0.34-0.69), respectively[7]
- This post hoc analysis evaluated the efficacy of ivonescimab plus chemotherapy on intracranial disease progression in HARMONi-A

OBJECTIVE

- To assess the effects of ivonescimab plus chemotherapy on intracranial PFS in patients with locally advanced or metastatic NSCLC in the HARMONi-A trial

METHODS

Study Design

- The HARMONi-A trial (NCT05184712) was a randomized, double-blind, placebo-controlled phase 3 trial in China, which included adult patients with locally advanced or metastatic, *EGFR* mutation–positive NSCLC whose disease had progressed after EGFR TKI therapy (**Figure 1**)[8]

Figure 1. Study Design

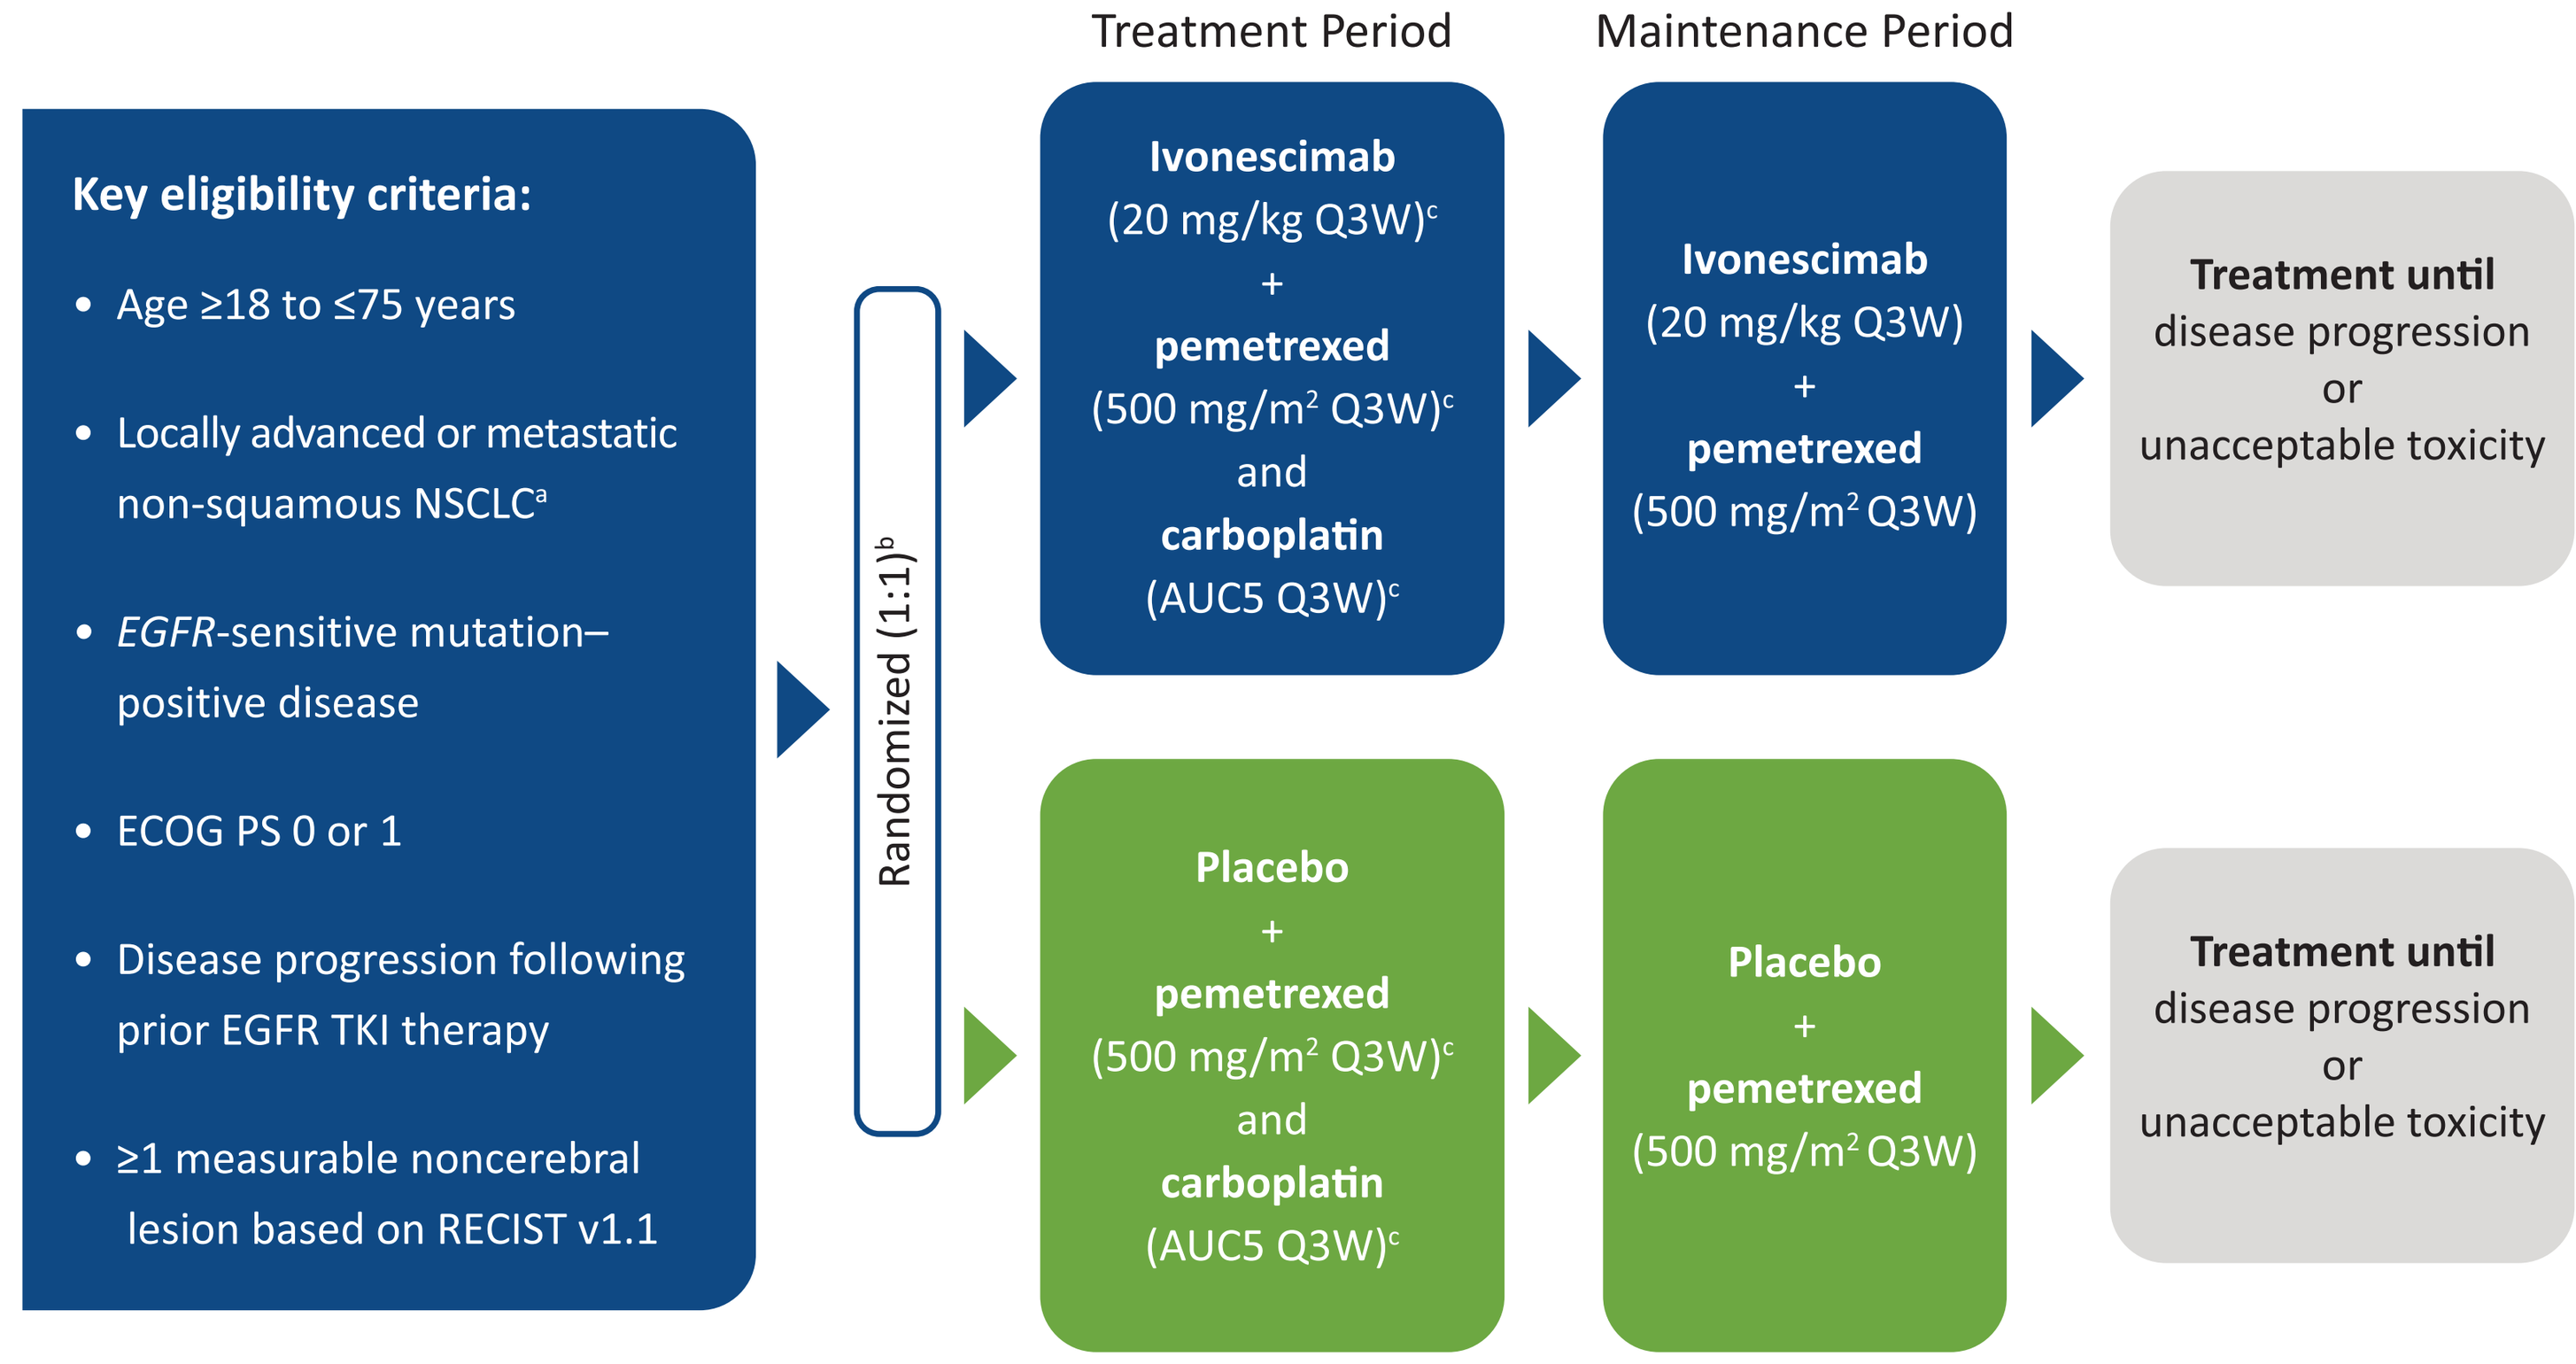

Treatment Period
Maintenance Period
Key eligibility criteria:
Age ≥18 to ≤75 years
Locally advanced or metastatic non-squamous NSCLC[a]
EGFR-sensitive mutation–positive disease
ECOG PS 0 or 1
Disease progression following prior EGFR TKI therapy
≥1 measurable noncerebral lesion based on RECIST v1.1
Randomized (1:1)[b]
Ivonescimab (20 mg/kg Q3W)[c] + pemetrexed (500 mg/m² Q3W)[c] and carboplatin (AUC5 Q3W)[c]
Ivonescimab (20 mg/kg Q3W) + pemetrexed (500 mg/m² Q3W)
Treatment until disease progression or unacceptable toxicity
Placebo + pemetrexed (500 mg/m² Q3W)[c] and carboplatin (AUC5 Q3W)[c]
Placebo + pemetrexed (500 mg/m² Q3W)
Treatment until disease progression or unacceptable toxicity

AUC5, area under the concentration time-curve value of 5 mg/mL/min; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; NSCLC, non–small cell lung cancer; Q3W, every 3 weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, version 1.1.; TKI, tyrosine kinase inhibitor.

[a]Histologically or cytologically confirmed non-squamous NSCLC (stage IIIB/C or stage IV; ineligible for surgery or local therapy).

[b]With stratification by exposure to third-generation EGFR TKIs (yes/no) and presence of brain metastases at baseline (yes/no).

[c]4 cycles (3 weeks/cycle).

Assessments and Outcomes

- Computed tomography or magnetic resonance imaging of the brain were performed for all patients at baseline
- In patients with brain metastases at baseline, tumor assessments via brain imaging were conducted every 6 weeks during the first 54 weeks and every 12 weeks thereafter
 - In patients without brain metastases at baseline, subsequent brain imaging was conducted at the discretion of the investigators on the basis of central nervous system symptoms
- The primary efficacy end point in the HARMONi-A trial was overall PFS by independent radiologic review committee (IRRC)
- For the current analysis, intracranial PFS was assessed by IRRC per Response Evaluation Criteria in Solid Tumors, version 1.1 in all patients enrolled (intention-to-treat [ITT] population) and in those with brain metastases at baseline
 - Intracranial PFS was defined as the time from randomization until intracranial progression or death from any cause in the absence of known intracranial progression
 - At the time of analysis, patients who were alive without intracranial disease progression were censored at their last evaluable scan

RESULTS

Participants

- As of March 10, 2023, a total of 322 patients were enrolled (161 in each arm) in the trial
 - The median duration of follow-up for all patients was 7.1 months (interquartile range [IQR], 5.4-9.0) for the ivonescimab plus chemotherapy arm and 8.2 months (IQR, 5.5-9.5) for the placebo plus chemotherapy arm
 - The median duration of exposure was 8.8 cycles (range, 1.0-16.9) and 7.3 cycles (range, 1.0-16.2) for ivonescimab and placebo, respectively
- Of the patients enrolled, 35 (21.7%) and 37 (23.0%) in the ivonescimab plus chemotherapy and placebo plus chemotherapy arms, respectively, had brain metastases at baseline (**Table 1**)
 - For patients with brain metastases at baseline, the median duration of follow up was 7.9 months (IQR, 7.1-8.4) and 8.2 months (IQR, 6.2-8.9) for the ivonescimab plus chemotherapy and placebo plus chemotherapy arms, respectively

Table 1. Baseline Characteristics

Characteristic	ITT population[a]		Patients with brain metastases at baseline	
	Ivonescimab plus chemotherapy n = 161	Placebo plus chemotherapy n = 161	Ivonescimab plus chemotherapy n = 35	Placebo plus chemotherapy n = 37
Age, median (range), years	59.6 (32.3-74.9)	59.4 (36.2-74.2)	59.2 (43.1-74.9)	57.7 (37.6-71.9)
Age ≥65 years, n (%)	50 (31.1)	51 (31.7)	10 (28.6)	11 (29.7)
Sex, n (%)				
Male	77 (47.8)	79 (49.1)	16 (45.7)	19 (51.4)
Female	84 (52.2)	82 (50.9)	19 (54.3)	18 (48.6)
ECOG PS, n (%)[b]				
0	24 (14.9)	34 (21.1)	7 (20.0)	7 (18.9)
1	137 (85.1)	127 (78.9)	28 (80.0)	30 (81.1)
Disease stage, n (%)[c]				
IIIB/C	3 (1.9)	5 (3.1)	0	0
IV	158 (98.1)	156 (96.9)	35 (100)	37 (100)
Patients with brain metastases, n (%)	35 (21.7)	37 (23.0)	35 (100)	37 (100)
Patients with liver metastases, n (%)	21 (13.0)	17 (10.6)	6 (17.1)	6 (16.2)
Patients with ≥3 distant metastases, n (%)	74 (46.0)	68 (42.2)	24 (68.6)	23 (62.2)
***EGFR* variant, n (%)**				
Exon 19 deletion	92 (57.1)	78 (48.4)	19 (54.3)	18 (48.6)
L858R	60 (37.3)	78 (48.4)	15 (42.9)	19 (51.4)
Other	35 (21.7)	25 (15.5)	7 (20.0)	6 (16.2)
Previous EGFR TKI therapy, n (%)				
1st/2nd generation TKI only	22 (13.7)	24 (14.9)	6 (17.1)	6 (16.2)
3rd generation TKI only	49 (30.4)	58 (36.0)	14 (40.0)	13 (35.1)
1st/2nd generation TKI, then 3rd generation TKI	90 (55.9)	79 (49.1)	15 (42.9)	18 (48.6)

ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; ITT, intention to treat; NA, not applicable; TKI, tyrosine kinase inhibitor.
All patients were enrolled in China, but race/ethnicity demographic information was not collected.

[a]Baseline characteristics for the patients in this analysis were previously published.[7]

[b]ECOG PS scores range from 0 to 5 (higher scores indicate greater disability).

[c]Stage at diagnosis was based on *AJCC Cancer Staging Manual*, 8th edition.[8]

Efficacy

- In the ITT population, the median intracranial PFS was not reached in either arm; HR was 0.39 (95% CI, 0.24-0.64; *P* < 0.001), favoring the ivonescimab plus chemotherapy arm
 - The 6-month PFS rates were 89.4% and 68.8% in the ivonescimab plus chemotherapy and placebo plus chemotherapy arms, respectively (**Figure 2**)
- In patients with brain metastases at baseline, median intracranial PFS was 8.4 months (95% CI, 7.0-NE) in the ivonescimab plus chemotherapy and 5.4 months (95% CI, 2.9-7.2) in the placebo plus chemotherapy arms; HR was 0.33 (95% CI, 0.15-0.74; *P* = 0.005), favoring the ivonescimab plus chemotherapy arm
 - The 6-month PFS rates were 83.7% and 42.9% in the ivonescimab plus chemotherapy and placebo plus chemotherapy arms, respectively (**Figure 3**)

Figure 2. Kaplan-Meier Curves of IRRC-Assessed Intracranial PFS (ITT Set)

	Ivonescimab + chemotherapy	Placebo + chemotherapy
No. of events	24	43
Median PFS (95% CI), months	NR (NE-NE)	NR (7.8-NE)
HR (95% CI)	0.39 (0.24-0.64; *P* <0.001)	

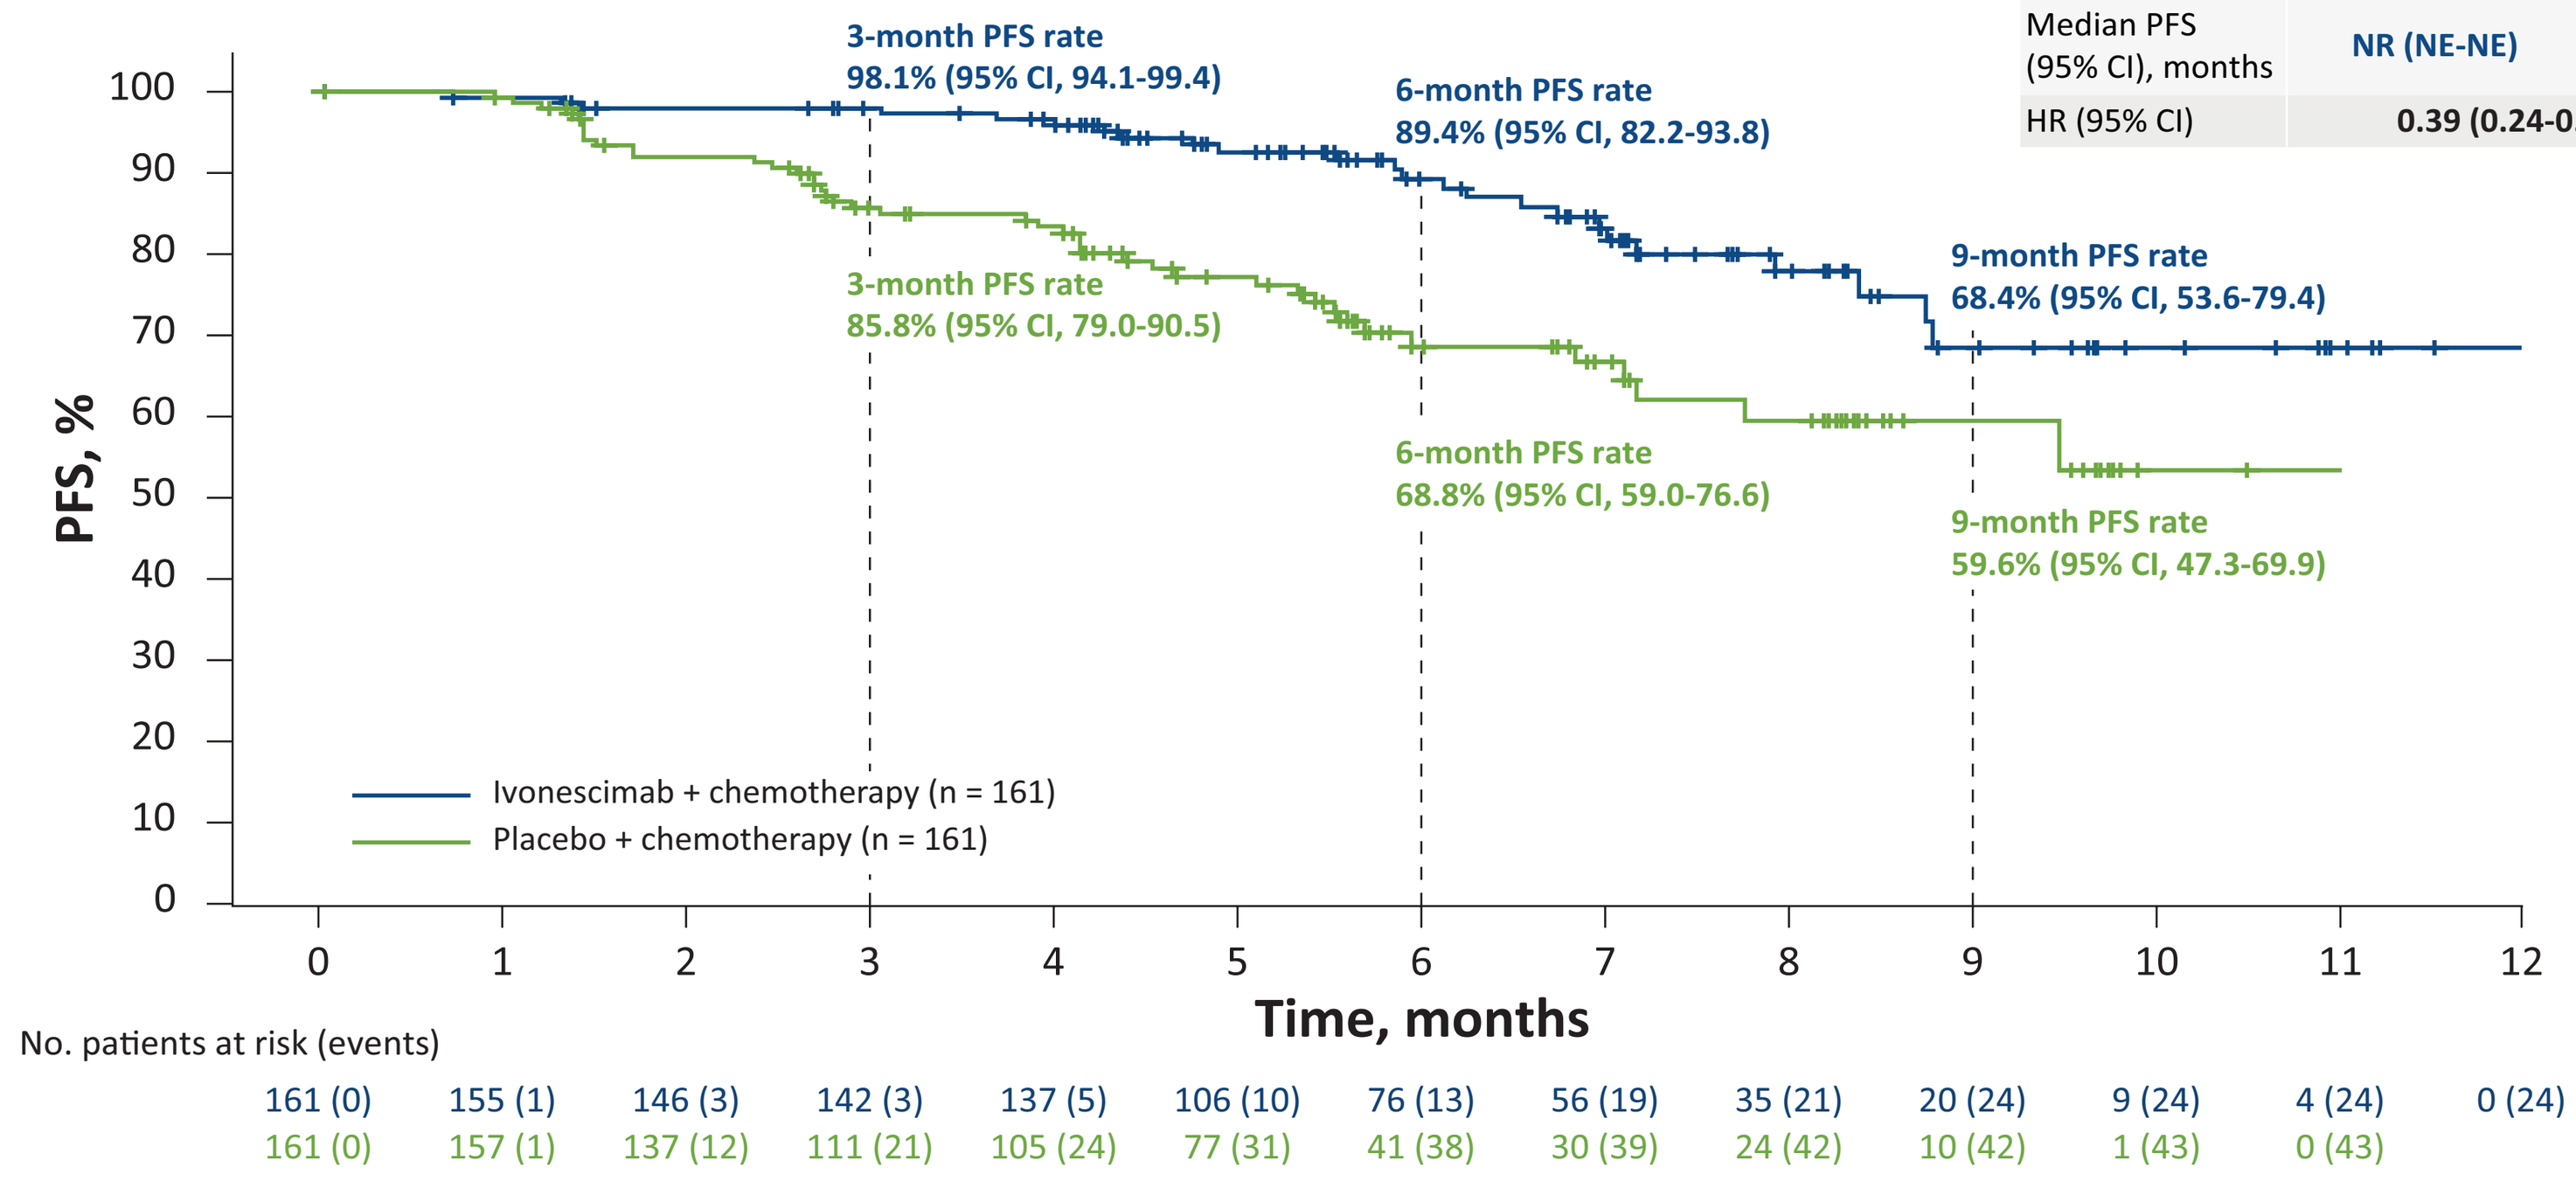

3-month PFS rate 98.1% (95% CI, 94.1-99.4)
6-month PFS rate 89.4% (95% CI, 82.2-93.8)
9-month PFS rate 68.4% (95% CI, 53.6-79.4)
3-month PFS rate 85.8% (95% CI, 79.0-90.5)
6-month PFS rate 68.8% (95% CI, 59.0-76.6)
9-month PFS rate 59.6% (95% CI, 47.3-69.9)
PFS, %
Time, months
Ivonescimab + chemotherapy (n = 161)
Placebo + chemotherapy (n = 161)
No. patients at risk (events)
161 (0) 155 (1) 146 (3) 142 (3) 137 (5) 106 (10) 76 (13) 56 (19) 35 (21) 20 (24) 9 (24) 4 (24) 0 (24)
161 (0) 157 (1) 137 (12) 111 (21) 105 (24) 77 (31) 41 (38) 30 (39) 24 (42) 10 (42) 1 (43) 0 (43)

HR, hazard ratio; IRRC, independent radiologic review committee; ITT, intention to treat; NE, not evaluable; NR, not reached; PFS, progression-free survival.

Figure 3. Kaplan-Meier Curves of IRRC-Assessed Intracranial PFS in Patients With Brain Metastases at Baseline

	Ivonescimab + chemotherapy	Placebo + chemotherapy
No. of events	9	19
Median PFS (95% CI), months	8.4 (7.0-NE)	5.4 (2.9-7.2)
HR (95% CI)	0.33 (0.15-0.74; *P* = 0.005)	

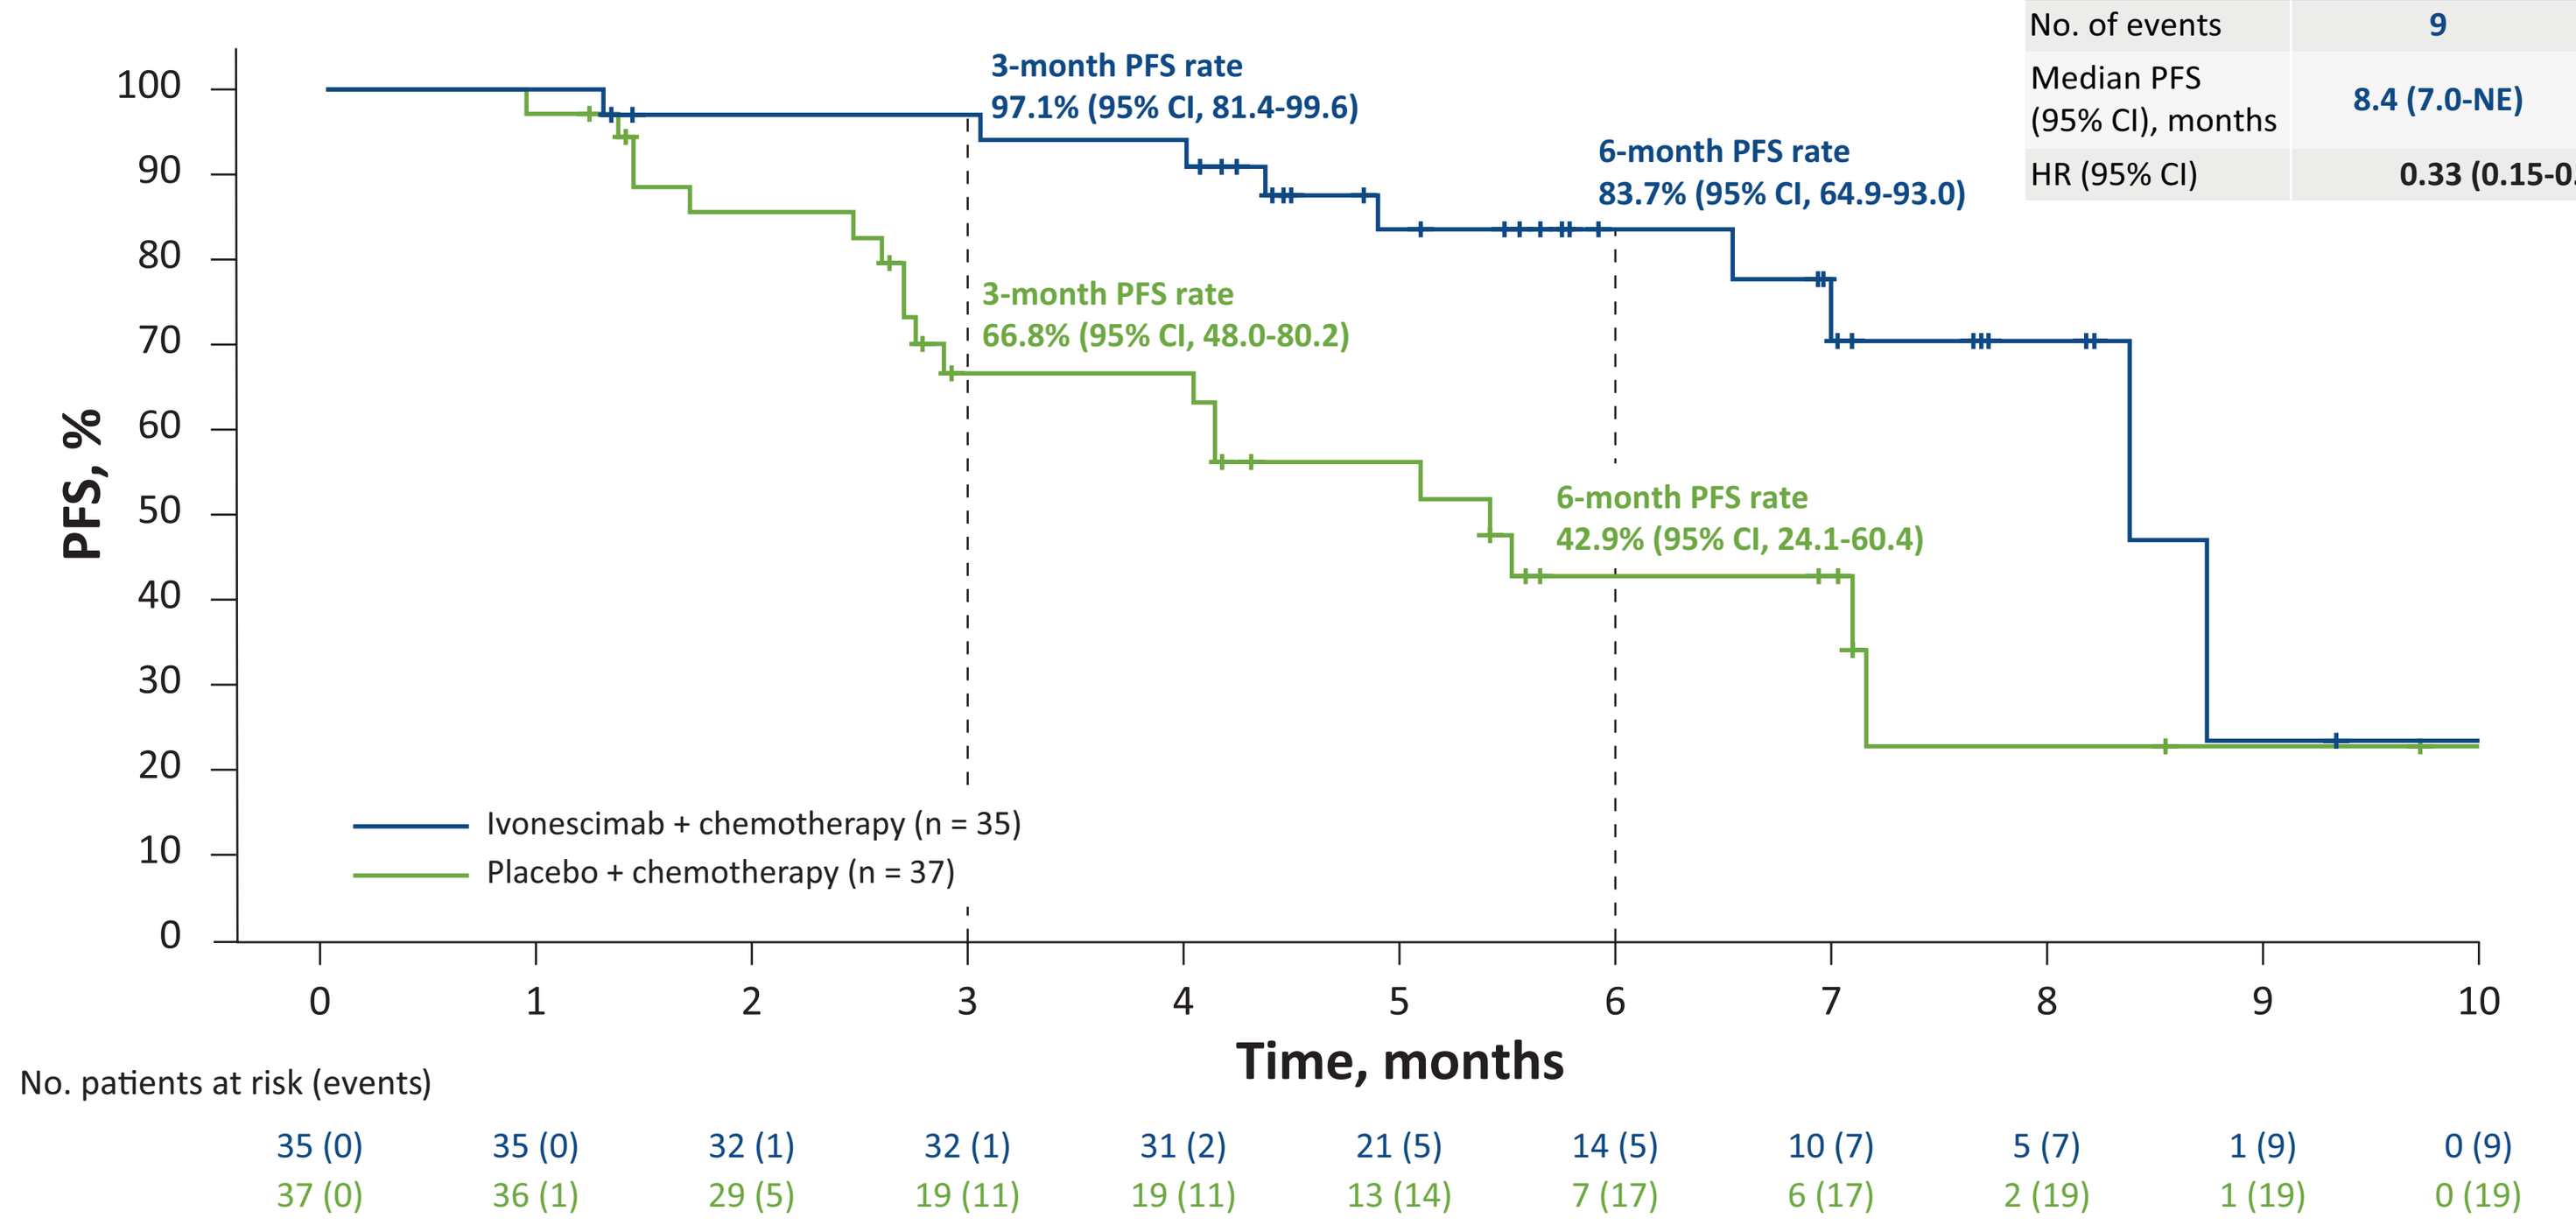

3-month PFS rate 97.1% (95% CI, 81.4-99.6)
6-month PFS rate 83.7% (95% CI, 64.9-93.0)
3-month PFS rate 66.8% (95% CI, 48.0-80.2)
6-month PFS rate 42.9% (95% CI, 24.1-60.4)
PFS, %
Time, months
Ivonescimab + chemotherapy (n = 35)
Placebo + chemotherapy (n = 37)
No. patients at risk (events)
35 (0) 35 (0) 32 (1) 32 (1) 31 (2) 21 (5) 14 (5) 10 (7) 5 (7) 1 (9) 0 (9)
37 (0) 36 (1) 29 (5) 19 (11) 19 (11) 13 (14) 7 (17) 6 (17) 2 (19) 1 (19) 0 (19)

HR, hazard ratio; IRRC, independent radiologic review committee; NE, not estimated; NR, not reached; PFS, progression-free survival.

CONCLUSIONS

- Ivonescimab combined with chemotherapy significantly improved intracranial PFS in patients with advanced *EGFR* mutation–positive NSCLC whose disease had progressed after previous EGFR-TKI therapy
- Results of this analysis support further evaluation of ivonescimab combined with chemotherapy for the treatment of *EGFR*-sensitive mutation–positive NSCLC

ACKNOWLEDGMENTS

The authors thank the participants and their caregivers, the investigators, and all the site staff who have been participating in this study. This study is sponsored by Akeso Biopharma, Inc. Medical writing assistance was provided by MEDiSTRAVA (San Francisco, CA, USA) and was funded by Summit Therapeutics.

REFERENCES

1. Riely GJ et al. *J Natl Comp Canc Netw*. 2024;22:249-274.
2. Hendriks LE et al. *Ann Oncol*. 2023;34:339-357.
3. Fu K et al. *J Hematol Oncol*. 2022;15:173.
4. Zhao Y et al. *EClinicalMedicine*. 2023;62:102106.
5. Wang L et al. *J Thorac Oncol*. 2024;19:465-474.
6. Frentzas S et al. *J Immunother Cancer*. 2024;12:e008037.
7. HARMONi-A study investigators. *JAMA*. 2024;332:561-570.
8. Amin MB et al. *CA Cancer J Clin*. 2017;67(2):93-99.

DISCLOSURES

Li Zhang has received research support from Hengrui, BeiGene, Xiansheng, Eli Lilly, Novartis, Roche, Hansoh, and Bristol Myers Squibb Pharma and consulting for MSD, Beigene, and Xiansheng Pharma.

Ivonescimab is an investigational therapy not approved by any regulatory authority other than China's National Medical Products Administration (NMPA).

CONTACT INFORMATION

Please contact the presenting author Dr Li Zhang at zhangli@sysucc.org.cn for questions or comments.

Copies of this poster obtained through QR, AR, and/or text key codes are for personal use only and may not be reproduced without permission of the authors.

By use of the QR Code, you agree to Summit's privacy notice, located at smmttx.com.

Presented at the 2024 ESMO Immuno-Oncology Annual Congress; December 11-13, 2024; Geneva, Switzerland